24 hour results from a US based, randomized, placebo-controlled, multi-centered trial, to evaluate a novel peppermint oil delivery system. Results from the IBSREST™ trial.

Michael S. Epstein, MD, AGAF, FACG[1]; Brooks D. Cash, MD, AGAF, FACG, FASGE[2]; Syed M. Shah, PhD[3]

1. Digestive Disorders Associates, Annapolis, MD, United States; 2. Division of Gastroenterology, University of South Alabama, Mobile, AL, United States; 3. IM HealthScience® LLC, Boca Raton, FL, United States

Introduction

Irritable bowel syndrome (IBS) is a chronic disorder with periodic exacerbations of symptoms that can be severe. The incidence of the3 major forms of IBS has been reported as IBS-M (61.0%), IBS-D (29.3%), and IBS-C (9.7%).1 While there are prescription options for IBS-C, there are no approved products for IBS-M and limited options, including restricted use alosetron, for IBS-D. The Irritable Bowel Syndrome Reduction Evaluation & Safety Trial (IBSREST) comprised of patients with IBS-M and IBS-D (in equal numbers) in order to address the needs of this large group of patients. Disturbance of small intestinal homeostasis, in particular transit time, has been implicated as a potential source of the clinical manifestations of IBS. IBgard® contains peppermint oil (PO), a proven ingredient to help normalize digestive and absorptive processes (including intestinal transit times), which are customarily disrupted by IBS. Along with the desired properties of PO's principal component L-menthol, in promoting uptake of important nutrients, it has been shown to possess anti-spasmodic, anti-inflammatory, serotonergic, and anti-bacterial properties. PO is available in the US as a medical food, as single-unit, liquid-filled, enteric-coated capsules. Such PO delivery methods are not specifically designed for targeted delivery to the small intestine, have a higher risk of dose-dumping, and generally reside longer in the stomach. As a result, patients receiving single-unit, enteric-coated PO can experience heartburn, nausea, and anal burning. IBgard® is a modern version of PO delivery consisting of triple-coated, sustained release microspheres designed to provide quick and reliable release of PO in the small intestine.

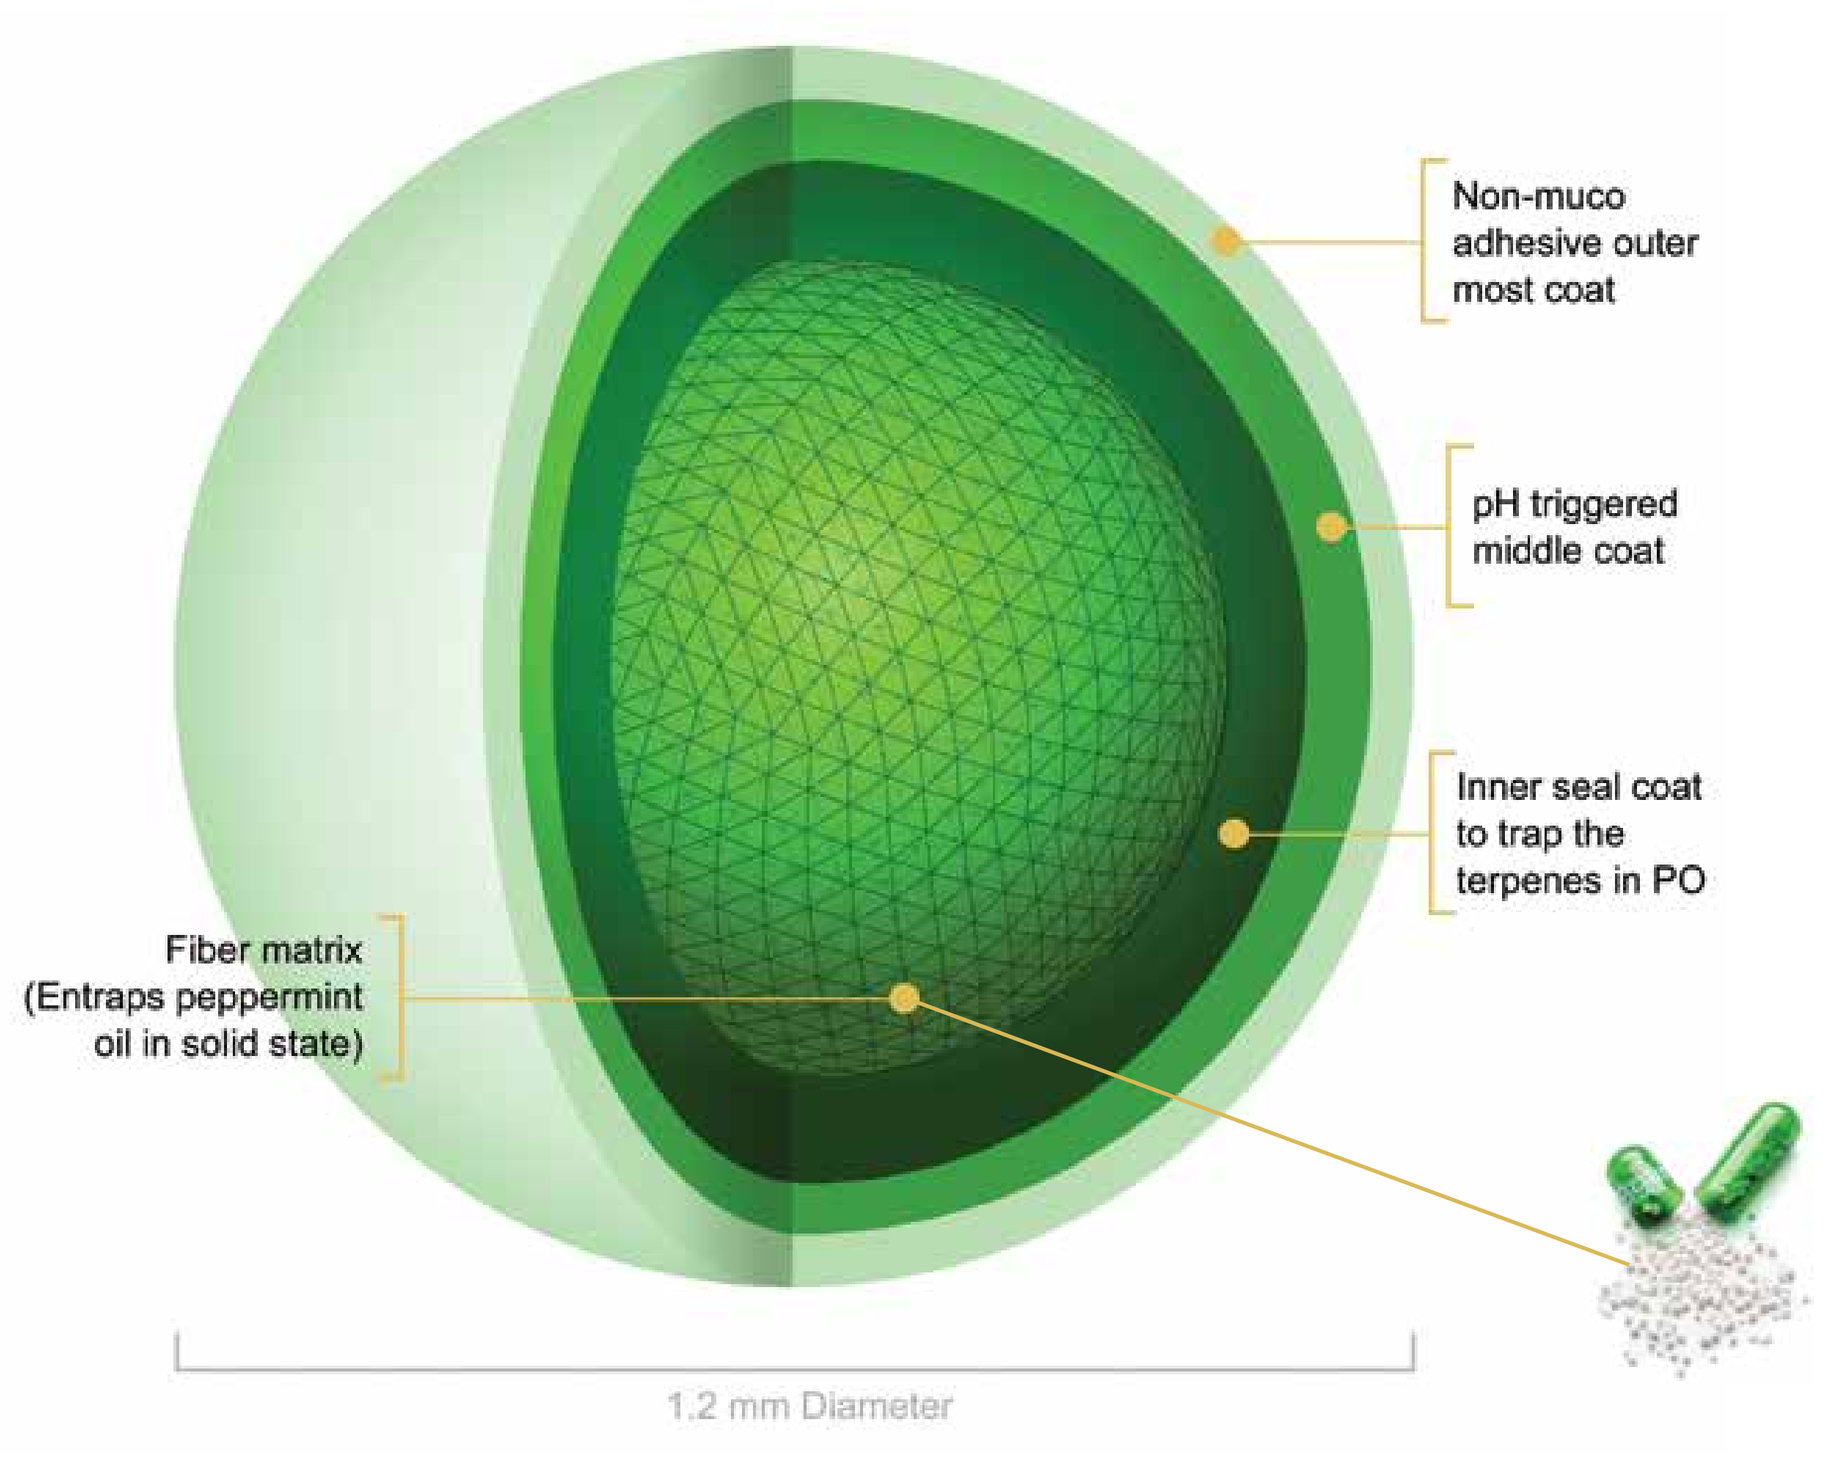
Non-muco adhesive outer most coat
pH triggered middle coat
Inner seal coat to trap the terpenes in PO
Fiber matrix (Entraps peppermint oil in solid state)
1.2 mm Diameter

Disclosures: MS Epstein, Chief Medical Advisor, IM HealthScience®, LLC; BD Cash, Consultant, IM HealthScience®, LLC; SM Shah, CIO, IM HealthScience®, LLC

© 2015 IM HealthScience™, LLC. Printed in USA IMHS19-15

IBSREST* Trial Objectives

Evaluate the effectiveness and safety of IBgard® for the management of IBS

- Confirm results of previous European clinical trials of PO in a U.S. population
- Determine if PO with Site Specific Targeting (SST®) technology results in rapid action and improved tolerability of PO in patients with IBS-M and IBS-D

* Irritable Bowel Syndrome Reduction Evaluation & Safety Trial

Methods

- Randomized to 2 IBgard capsules (90 mg PO) or identical placebo TID 30-90 min. before meals
 - Rescue medications were not allowed during the study
- Before enrollment, patients underwent testing to exclude organic disease and a 2-week washout period of all prohibited medications
- Patients maintained a 2-week baseline diary with daily assessments of bowel movements and IBS symptoms
- Prior to and 24 hours after randomization, measurements of IBS symptom frequency and intensity (both on a scale of 0-4) were obtained
 - IBS symptoms included abdominal pain or discomfort, abdominal bloating or distention, pain at evacuation, urgency of bowel movement, constipation, diarrhea, passage of mucus or gas, and a sense of incomplete evacuation.
 - Methods and endpoints (TISS) approximated those of Cappello et al.[2]

Inclusion Criteria

- Females and males 18-60 years of age
- Met Rome III IBS criteria for IBS-M or IBS-D
- Average daily IBS-related abdominal pain ≥4.0 on a 0 to 10 scale each week of 2 week baseline period
- TISS ≥2.0 on a 0 to 4 scale
- Subject was not planning to change his/her usual diet and lifestyle during the course of the study

Exclusion Criteria

- Diagnosis of IBS-C or IBS-U
- Organic gastrointestinal disease
- Refusal to discontinue one or more prohibited medications for at least 7 days prior to beginning the baseline diary and throughout the remainder of the study

Results

Baselines reported in all tables and figures were not significantly different between IBgard and placebo (*P*> 0.05).

Table 1. Demographic Characteristics

	IBgard® n (%)	Placebo n (%)
n	35	37
Mean Age (years)	40.2	41.1
Gender		
Female	28 (80.0)	26 (70.3)
Male	7 (20.0)	11 (29.7)
Race		
Caucasian	29 (82.9)	27 (73.0)
African American	6 (17.1)	8 (21.6)
Asian	0	1 (2.7)
Other	0	1 (2.7)
IBS Subtype		
IBS-M	16 (45.7)	18 (48.6)
IBS-D	19 (54.3)	19 (51.4)

Figure 1. Reduction in Total IBS Symptom Score at 24 Hours*

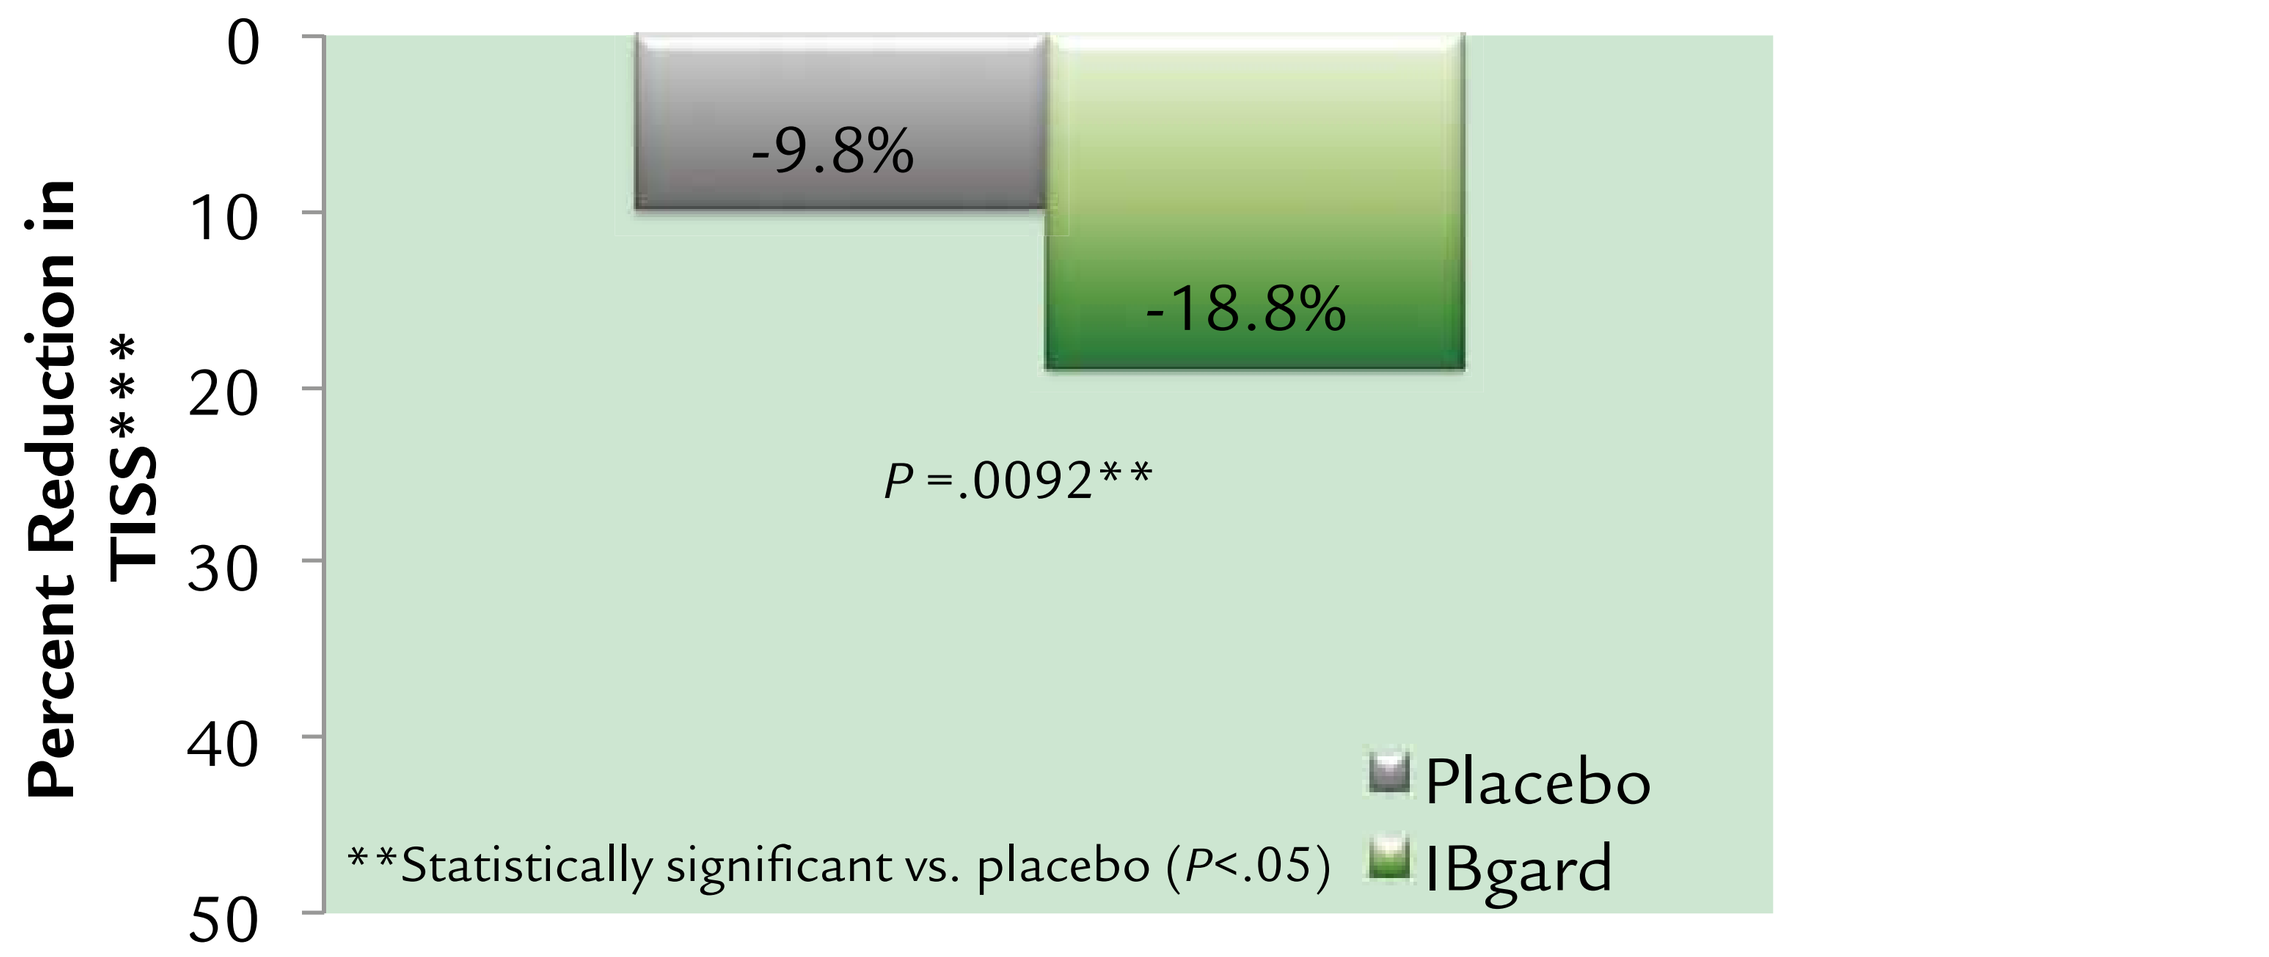
Percent Reduction in TISS***
0
10
20
30
40
50
-9.8%
-18.8%
P =.0092**
Placebo
IBgard
**Statistically significant vs. placebo (P<.05)

*Percent reduction from baseline.
***TISS=mean intensity and frequency score for each IBS symptom summed and divided by 8

Figure 2. Reduction in All 8 Symptoms at 24 Hours

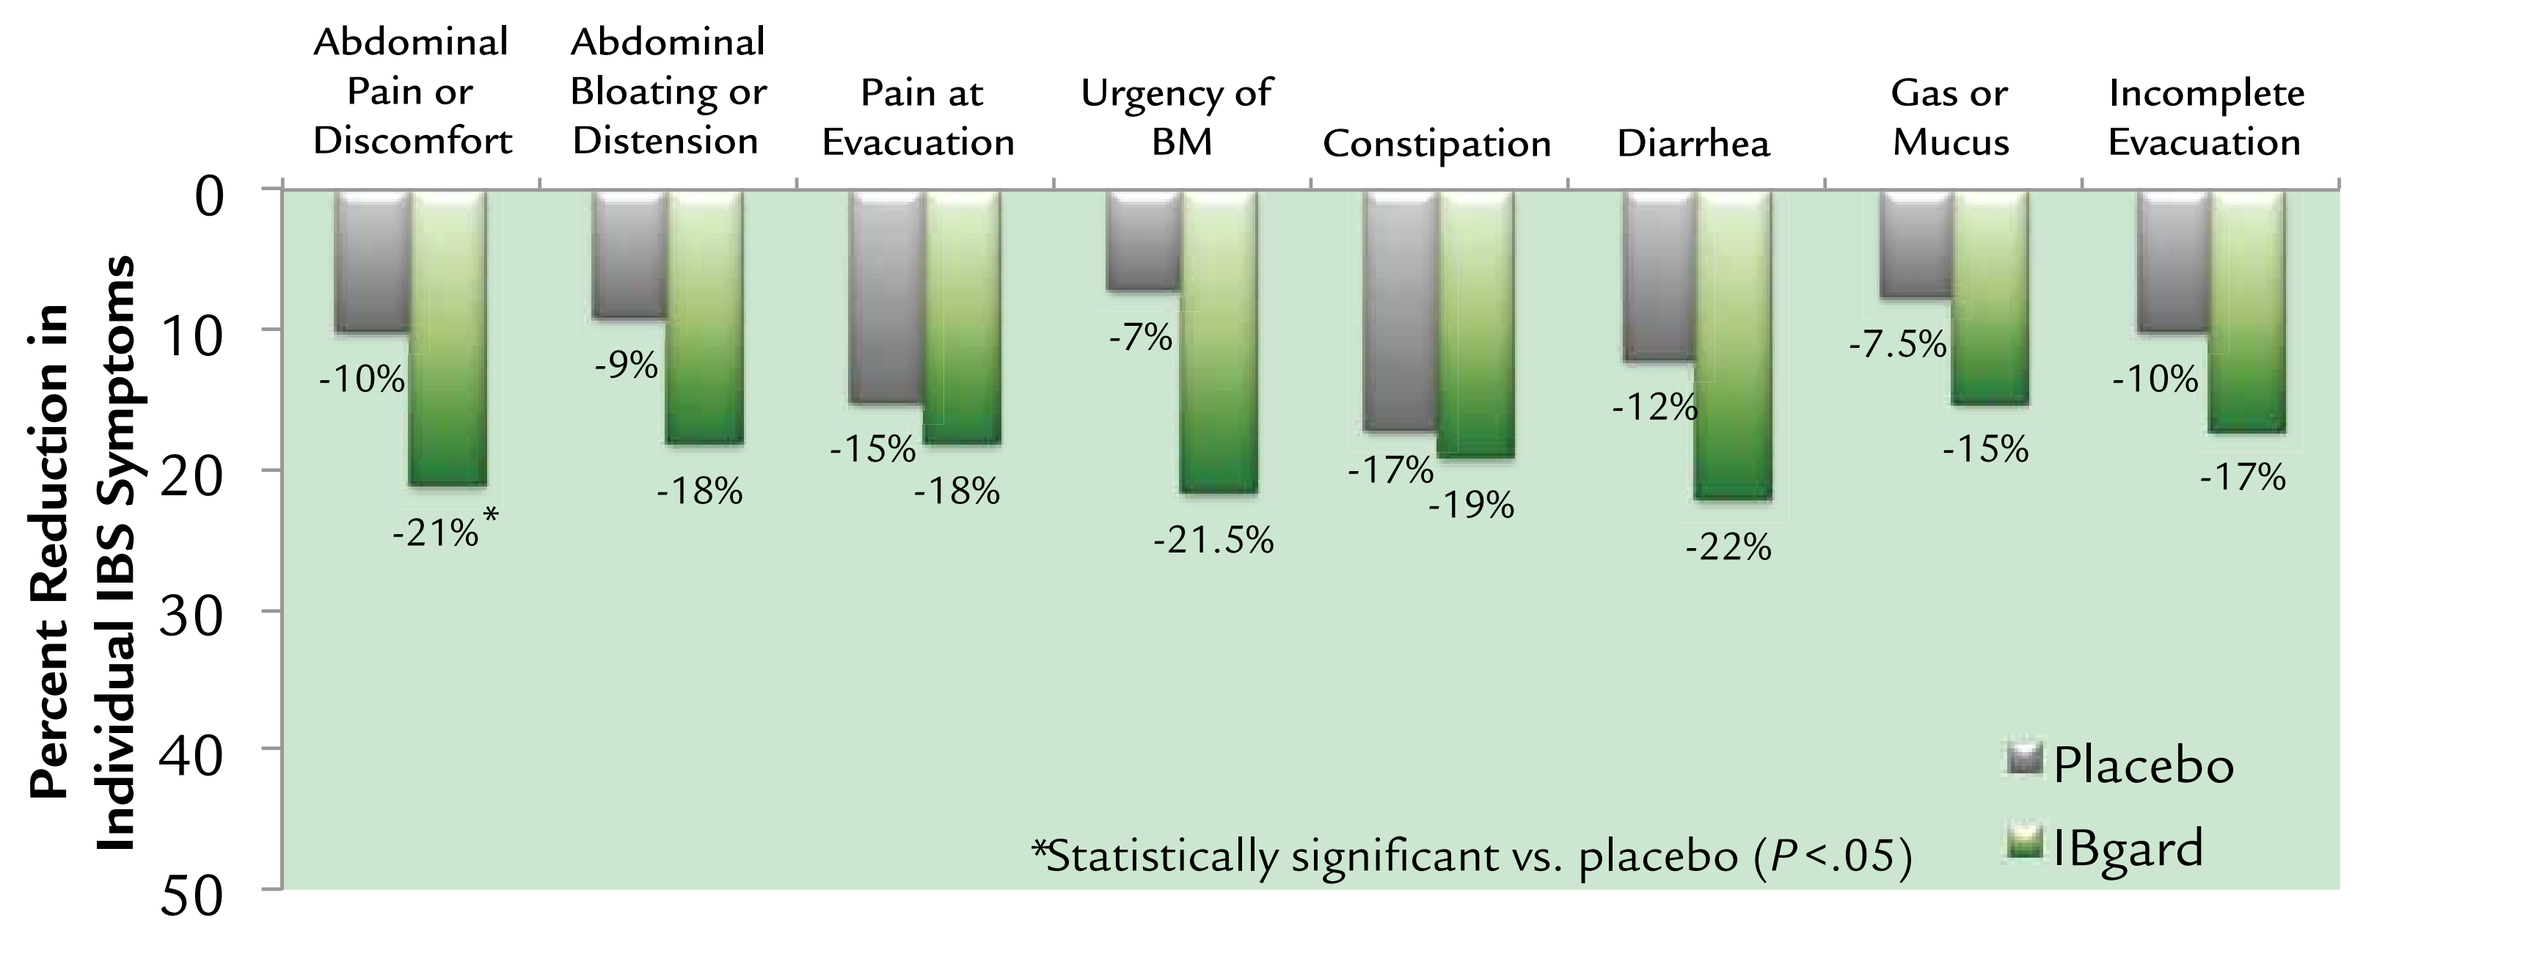
Abdominal Pain or Discomfort
Abdominal Bloating or Distension
Pain at Evacuation
Urgency of BM
Constipation
Diarrhea
Gas or Mucus
Incomplete Evacuation
Percent Reduction in Individual IBS Symptoms
0
10
20
30
40
50
-10%
-21%*
-9%
-18%
-15%
-18%
-7%
-21.5%
-17%
-19%
-12%
-22%
-7.5%
-15%
-10%
-17%
Placebo
IBgard
*Statistically significant vs. placebo (P<.05)

Percent reduction from baseline in individual IBS symptoms (average of frequency and intensity)

Figure 3. Reduction in abdominal pain and discomfort at 24 hours

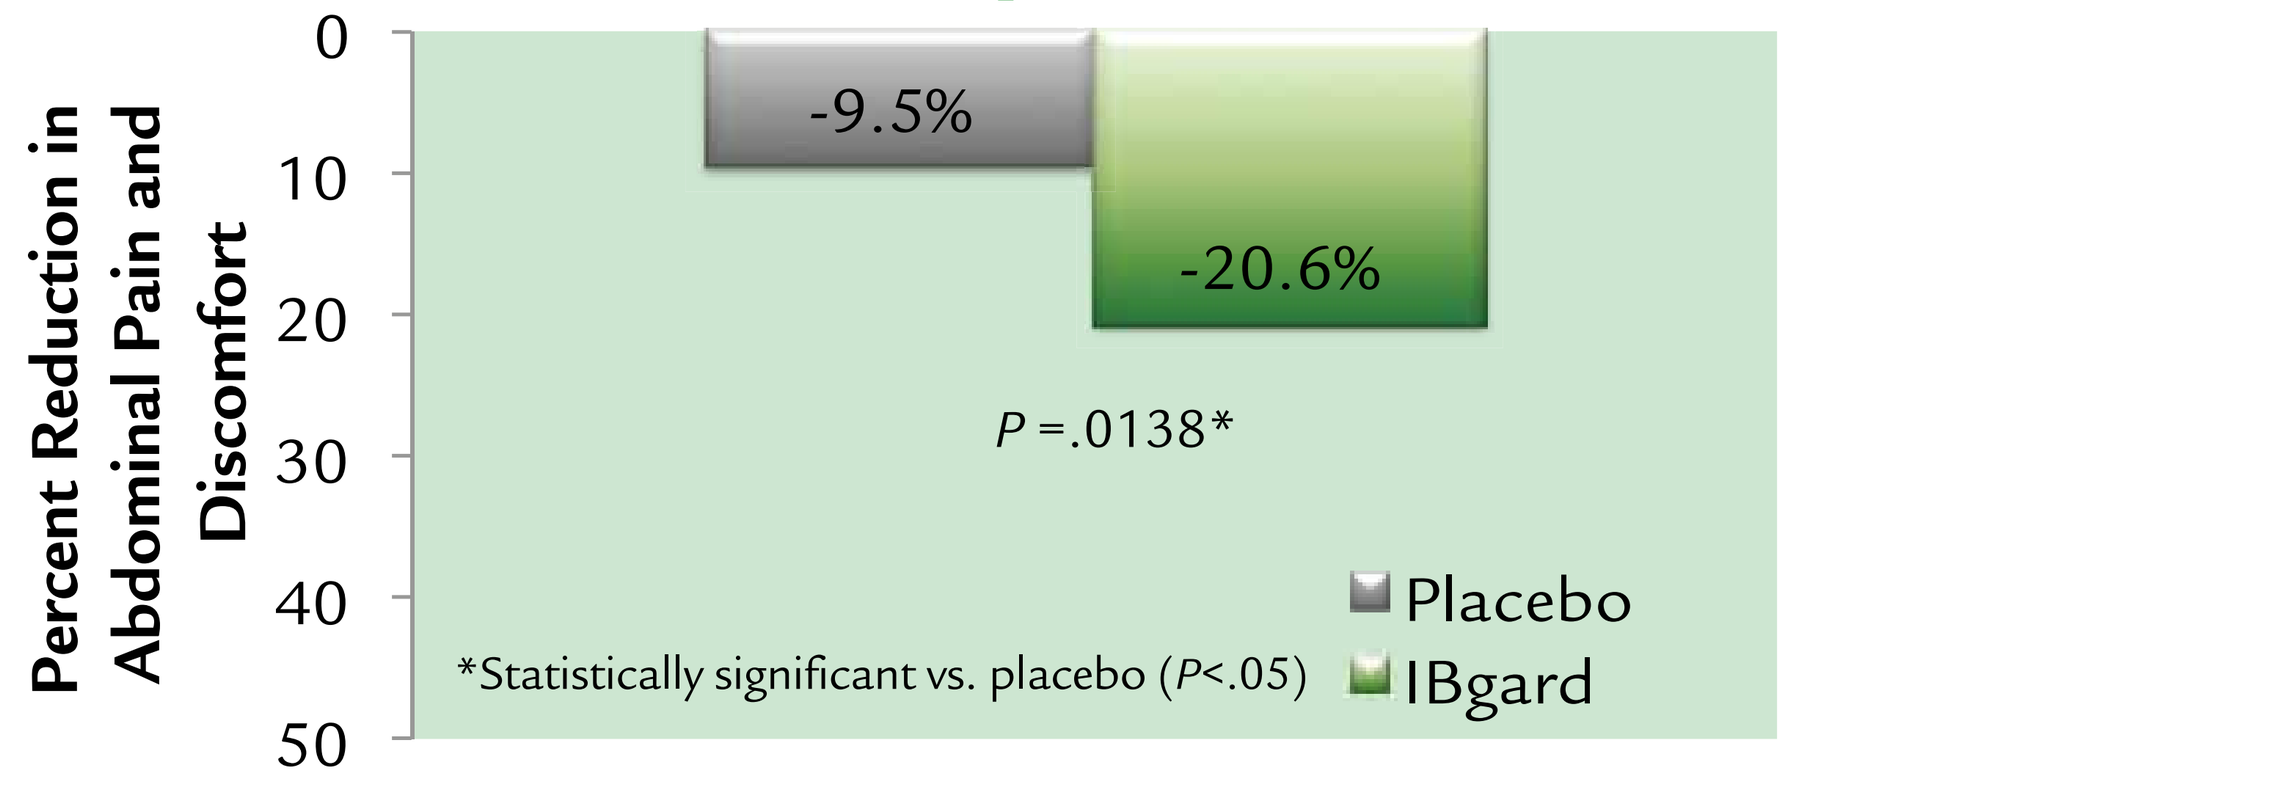
Percent Reduction in Abdominal Pain and Discomfort
0
10
20
30
40
50
-9.5%
-20.6%
P =.0138*
Placebo
IBgard
*Statistically significant vs. placebo (P<.05)

Percent reduction from baseline in abdominal pain and discomfort (average of frequency and intensity)

Figure 4. Reduction in Intensity of Bowel Movement Urgency at 24 Hours

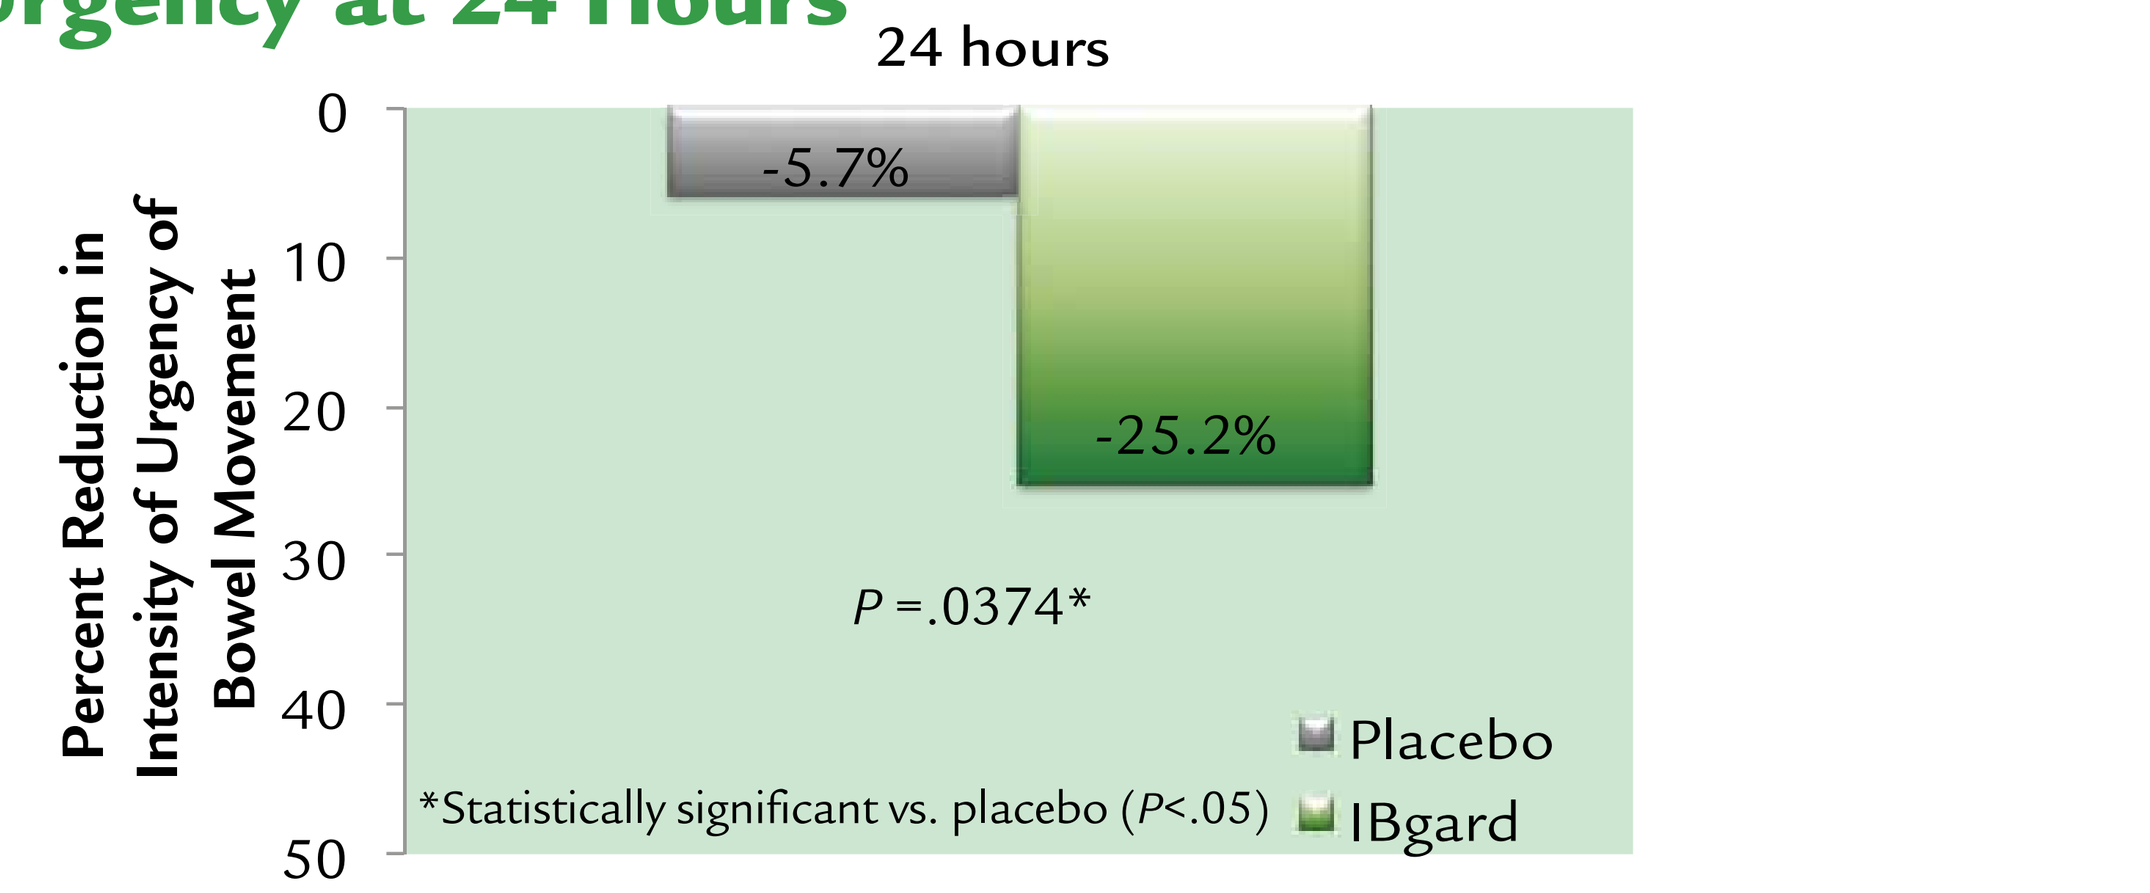
24 hours
Percent Reduction in Intensity of Urgency of Bowel Movement
0
10
20
30
40
50
-5.7%
-25.2%
P =.0374*
Placebo
IBgard
*Statistically significant vs. placebo (P<.05)

Percent reduction from baseline in the mean intensity of urgency of bowel movement

Conclusions

- In the IBSREST Trial, measurements of all 8 IBS symptoms (average of intensity and frequency) trended in favor of IBgard, as compared to placebo, within 24 hours of the first dose, and abdominal pain and discomfort reached statistical significance
- Subjects in the IBgard treated group experienced statistically significant decreases from baseline, compared to the placebo group, at 24 hours, in the following:
 - TISS (total IBS symptom score)
 - Abdominal pain and discomfort (average of frequency and intensity)
 - Urgency of bowel movements (intensity)
- IBgard was well tolerated and associated with no treatment emergent adverse events during the first 24 hours, which was comparable to placebo.

References

1. Drossman et al. *J. Clin. Gastroenterol*. 2009;43(6) 541-550.
2. Cappello et al. *Dig Liv Dis.* 2007;39:530-6.

Acknowledgements: Principal Investigators on the trial: Dennis S Riff, MD, FACG, CPI; Steven C Bowman, MD; Gigi Claire Lefebvre, MD; and Richard Krause, MD. Palm Beach CRO, LLC helped conduct the trial. SDC Biostatistics and Data Management provided power and statistical analyses. Editorial support was provided by Premier Healthcare and Precise Medical Writing, LLC. Design support was provided by Skylographic Design, LLC. The clinical study report was prepared by Hubbell Consulting, LLC.